PAIN: 712

Rapid pain relief with fentanyl citrate nasal spray (Nasalfent®) in cancer patients with breakthrough pain

Authors:
Davis G[1], Knight AC[2], Love R[3], Fisher AN[4]

1. St Joseph's Hospital, Thunder Bay, Ontario, Canada
2. Régional de Sudbury Regional Hospital, Ontario, Canada
3. Nanaimo Regional General Hospital, British Columbia, Canada
4. Archimedes Development Ltd, Nottingham UK

Introduction

- Cancer is a substantial cause of severe pain. Patients may suffer with two manifestations of pain. Background pain, a relatively constant pain, and breakthrough cancer pain, intermittent episodes of pain superimposed on the background pain.
- Breakthrough cancer pain is often unpredictable and is characterised by a rapid onset of intense pain that may last for a short time (usually <45 minutes). It is relatively widespread, reported in up to 95% of cancer patients.[1]
- Treatment for breakthrough cancer pain requires rapid onset of pain relief (ideally within five minutes), and a short duration of action (around 30-40 minutes).
- Convenience and ease of administration for patients are also important considerations.
- Fentanyl, a potent opioid with a short duration of action and an established side-effect profile, is approved to be administered through a variety of routes for the alleviation of severe pain and is particularly suited for breakthrough cancer pain.
 - o Additional routes of delivery of fentanyl are currently being investigated; the nasal route has many advantages over other routes including speed of delivery and patient acceptability.
- A Phase I study has demonstrated fentanyl citrate nasal spray (FCNS [Nasalfent®]), a novel formulation of fentanyl using the proprietary pectin system, PecSys™, has a pharmacokinetic profile highly suited to the natural time-course of the typical breakthrough pain episode.
- PecSys, a simple aqueous solution that forms a gel on contact with calcium ions in the nasal mucosa, optimises fast delivery of lipophilic molecules to the highly vascularised nasal mucosa, enhancing drug performance and patient acceptability.
- This Phase II trial was conducted to explore the efficacy and safety of Nasalfent in the treatment of breakthrough pain.
- A further aim of the study was to determine the acceptability to patients of Nasalfent in the treatment of breakthrough pain.
- The safety and tolerability results of this study are reported separately.[2]

Methods

Patients

- This study was an open-label, multi-centre, inpatient study.
- Inclusion criteria included:
 - o Pain due to cancer requiring the use of strong opioids regularly and as required to relieve episodes of breakthrough pain.
 - o Breakthrough pain of at least moderate severity (pain score ≥2; 5-point scale).
 - o Breakthrough pain responding to opioids.
- Exclusion criteria included:
 - o Too frail or unwell to participate.
 - o Pain that responds poorly to opioids.
 - o Patients with a known intolerance or allergy to opioids.
 - o Nasal passages occluded or congested.
 - o Patients with a history of nasal pathology including polyps or nasal obstructions, at the time of enrolment.

Study design

- The study was conducted in two parts:
 - o Part 1, dose titration: 18 patients were titrated in a dose escalation sequence of Nasalfent (25mcg-800mcg) to identify an effective dose for up to a maximum of 3 episodes of breakthrough pain.
 - o Part 2, efficacy assessment: 15 patients utilised their effective dose as identified in Part 1, for up to 4 episodes of breakthrough pain.
- The primary endpoint of the study was effective relief of breakthrough pain as indicated by:
 - o Reduction in the pain intensity score (assessed by the patient using a 5-point (0–4) pain descriptor scale).
 - o Onset of pain relief (assessed using a 9-point (-4 to 4) scale).
 - o Time to meaningful pain relief.
- Safety endpoints are reported elsewhere.[2]

Results

Patient demographics

- 29 patients consented to enter the study; 23 entered Part 1 and 18 patients successfully completed this phase.
- During Part 1, 3 patients obtained no pain relief at the highest permitted dose of Nasalfent (800mcg). Therefore, 15 patients entered Part 2 and were treated and assessed for up to 4 breakthrough pain episodes.

Efficacy Population

- 55 episodes of breakthrough pain, in 15 patients, were included in the efficacy analysis.
- Statistically significant improvements from baseline in pain intensity (pain intensity difference, PID) and the onset of pain relief (PR) were seen within 5 minutes after dosing (both $p<0.05$; Figures 1 and 2).

Figure 1:
Mean (±SD) Patient Pain Intensity Difference compared with baseline* (Population: Efficacy)

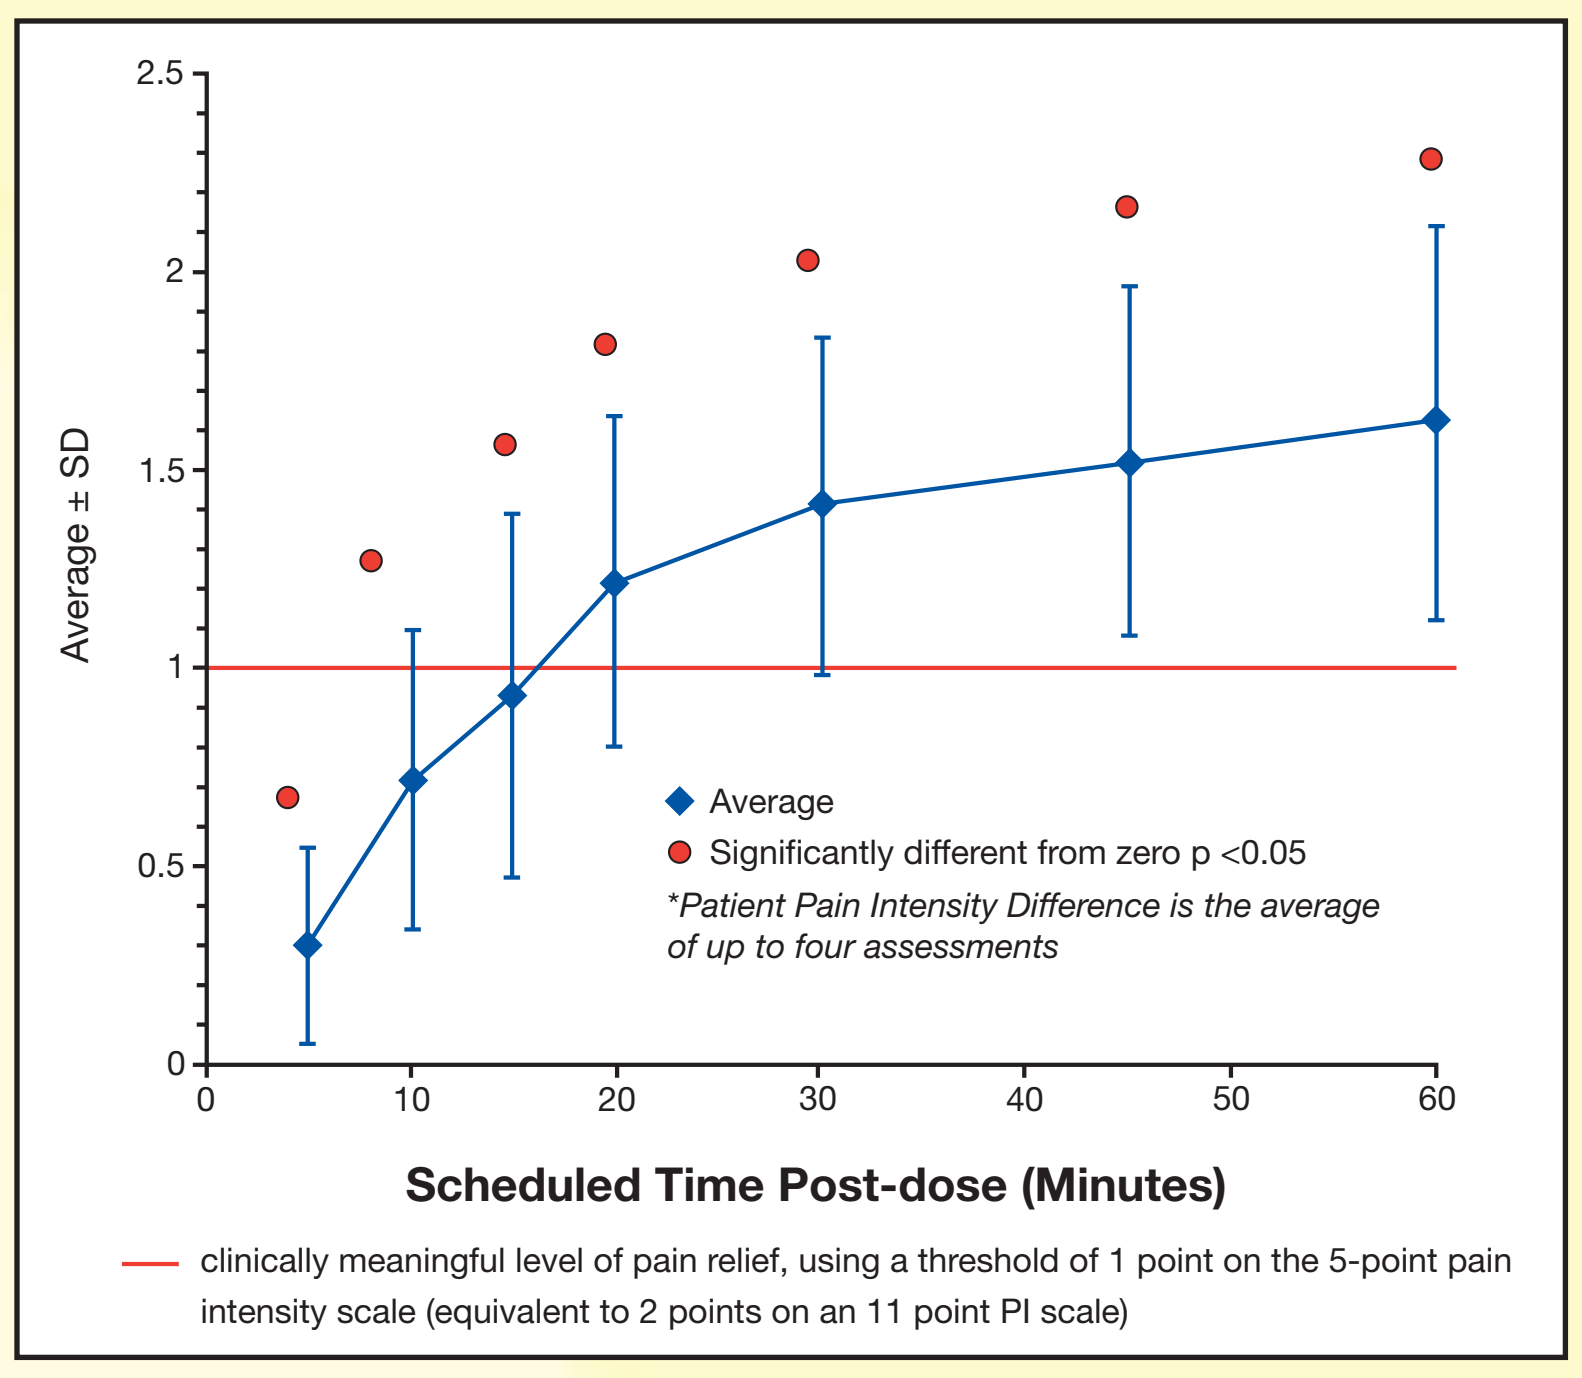
Average ± SD
Average
Significantly different from zero p <0.05
*Patient Pain Intensity Difference is the average of up to four assessments
Scheduled Time Post-dose (Minutes)
clinically meaningful level of pain relief, using a threshold of 1 point on the 5-point pain intensity scale (equivalent to 2 points on an 11 point PI scale)

Figure 2:
Mean (±SD) Patient Pain Relief Scores compared with baseline* (Population: Efficacy)

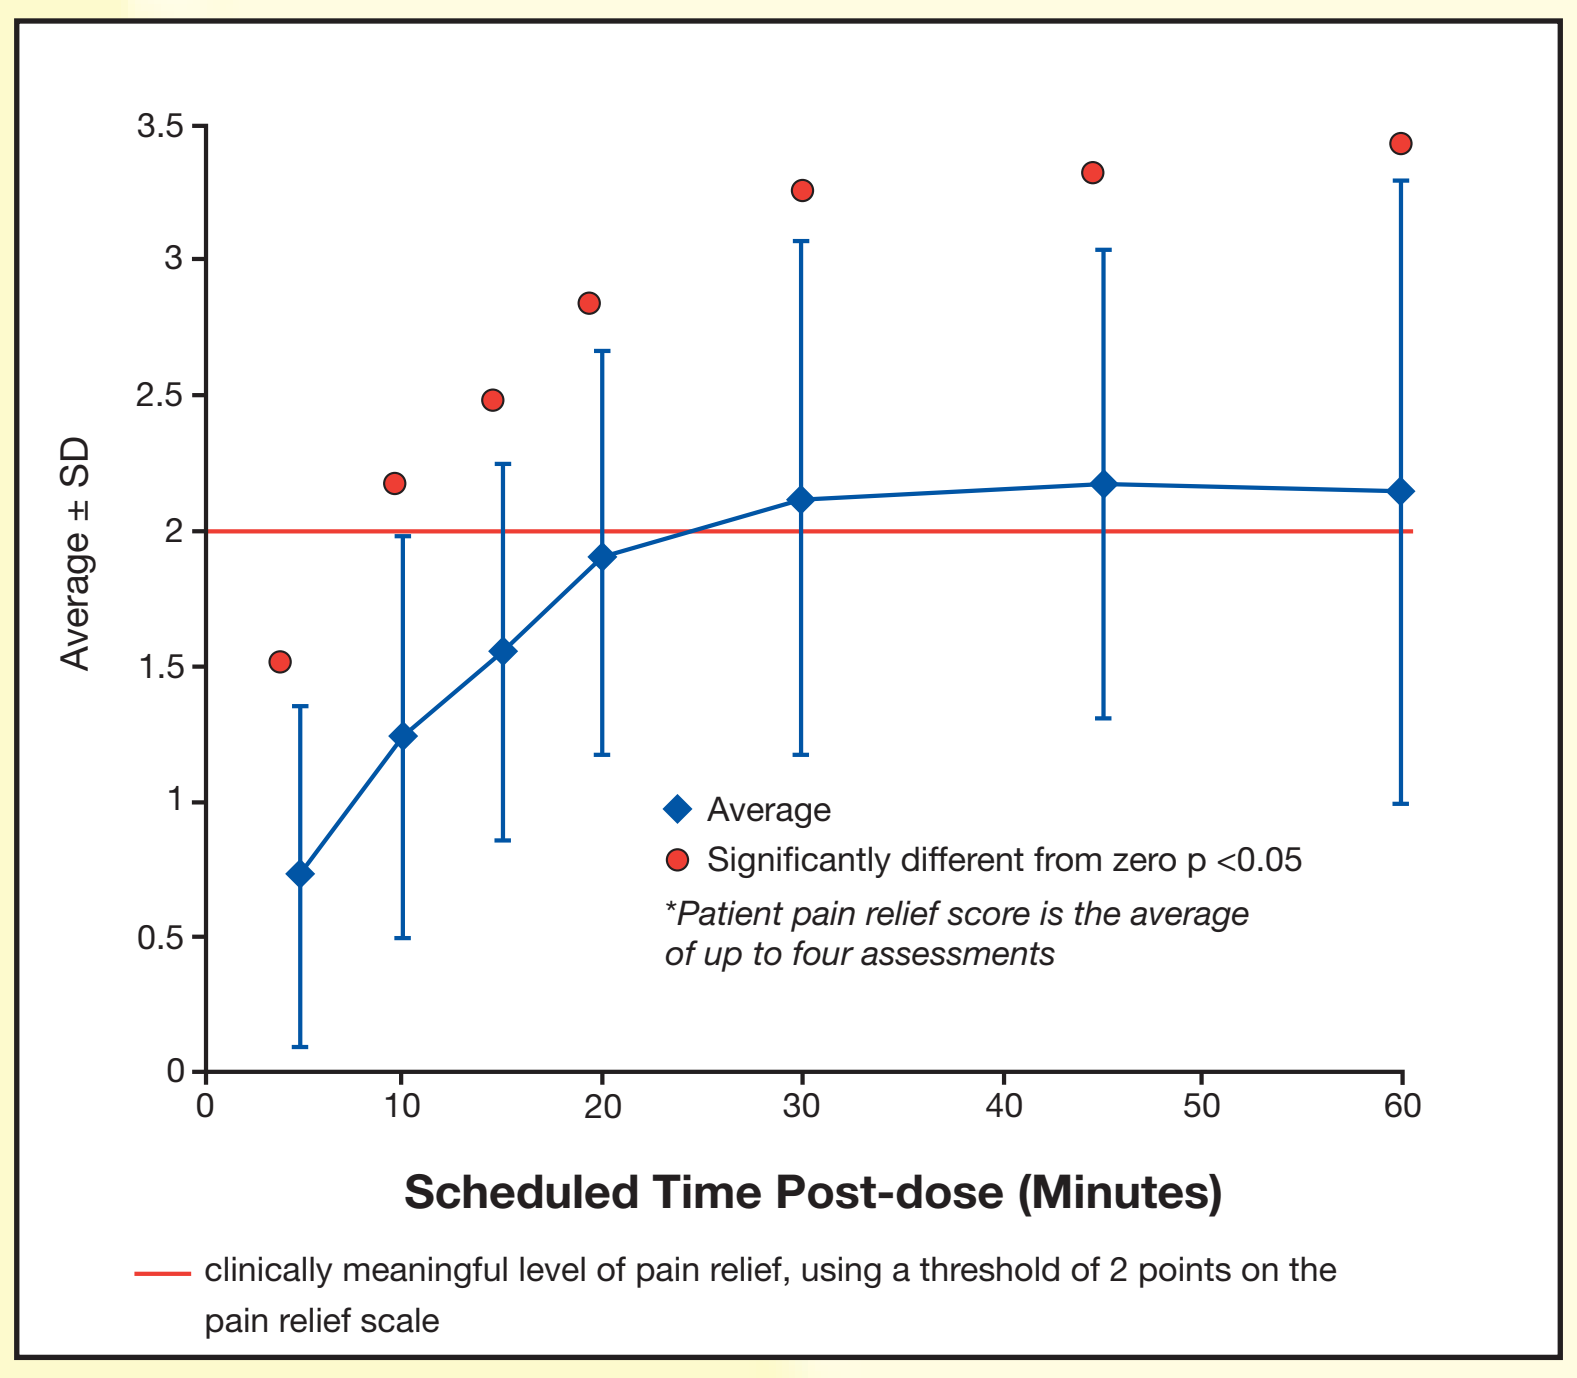
Average ± SD
Average
Significantly different from zero p <0.05
*Patient pain relief score is the average of up to four assessments
Scheduled Time Post-dose (Minutes)
clinically meaningful level of pain relief, using a threshold of 2 points on the pain relief scale

- Mean change in PID and mean PR score for patients who identified an effective dose reached a clinically meaningful level approx 16 minutes and 24 minutes after dosing, respectively.
- All patients experienced at least one episode with meaningful PR, and 96% (53 of the 55) of breakthrough pain episodes recorded some degree of pain relief.
- Of more clinical relevance are the percentages of treated episodes that saw clinically significant pain relief at each time point after treatment (The most widely accepted standard for clinically meaningful pain relief is a 33% fall in baseline pain intensity; Figure 3).

Figure 3:
Percentage of Episodes with Clinically Meaningful Pain Relief*

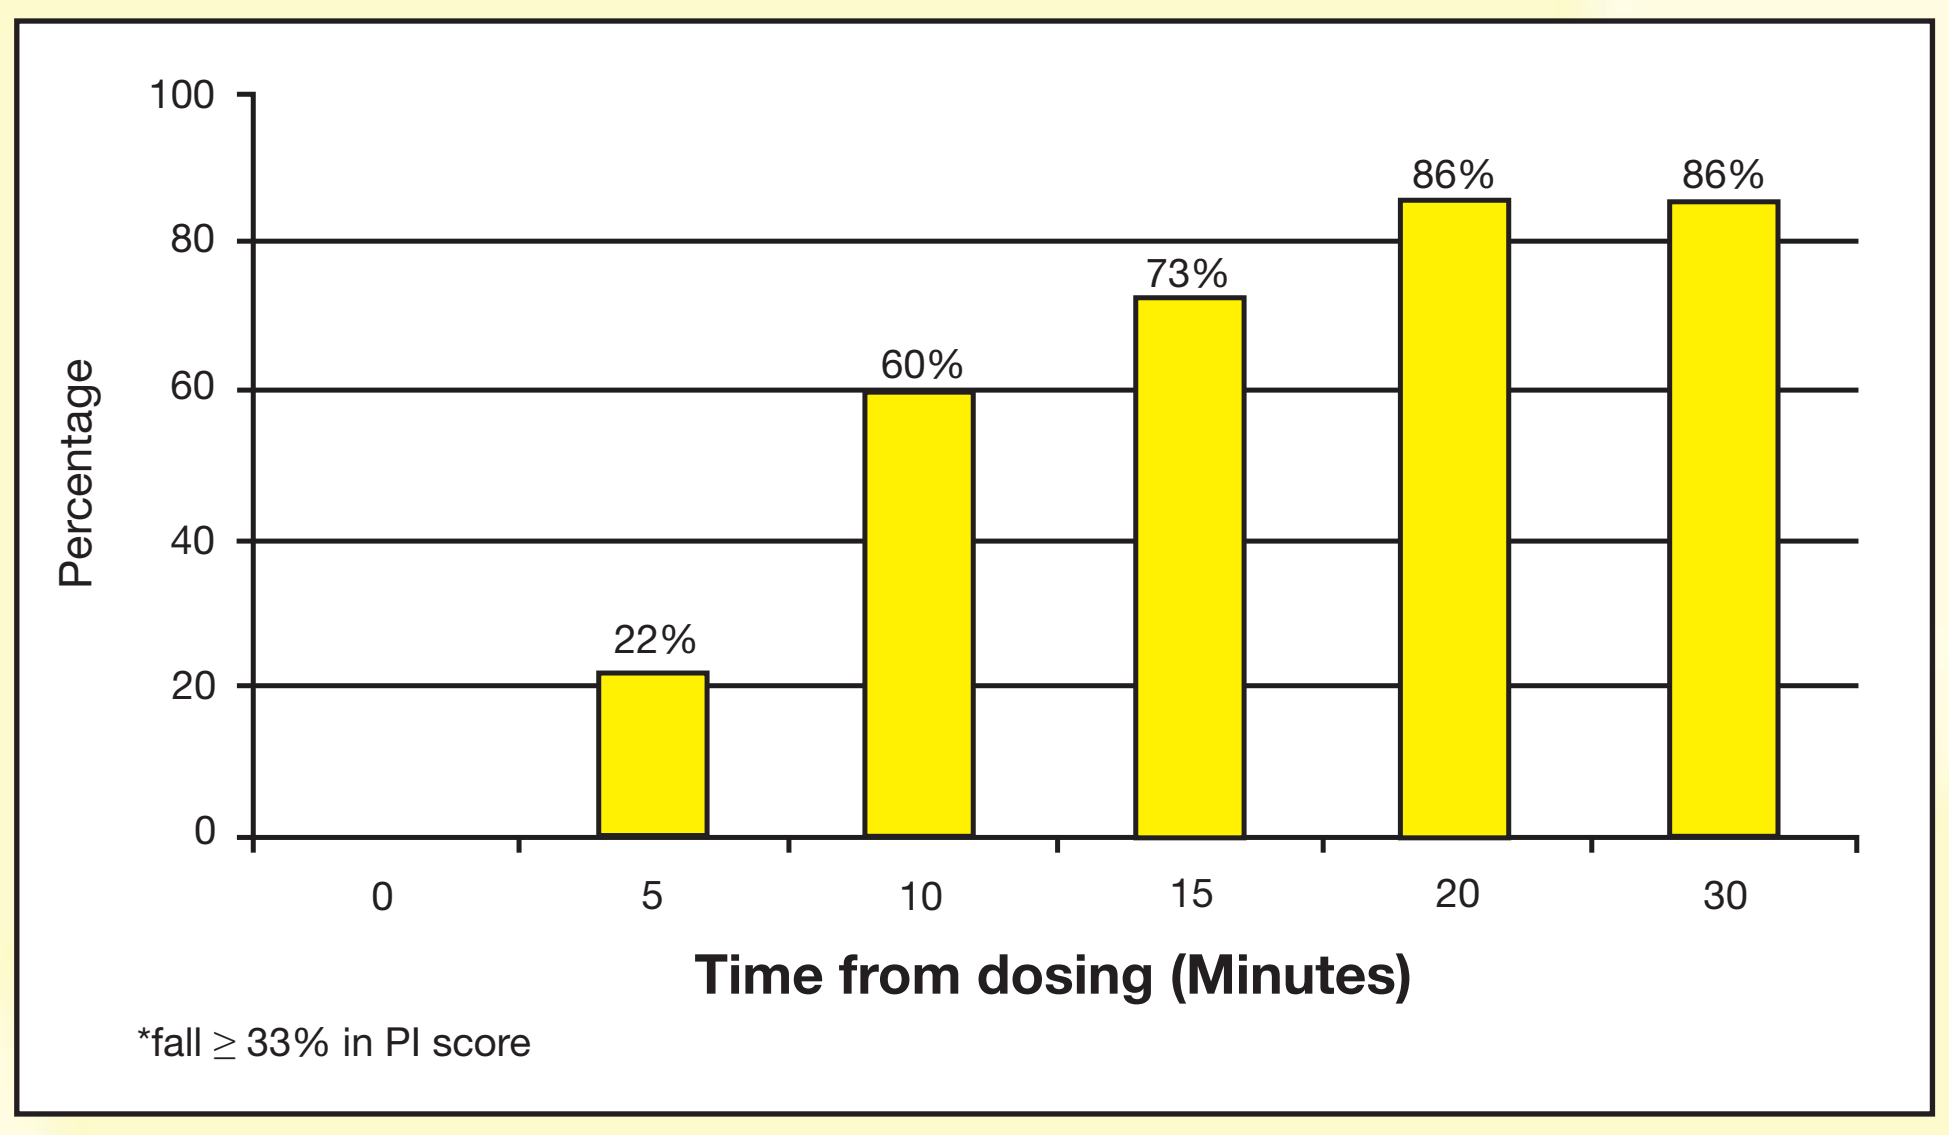
Percentage
22%
60%
73%
86%
86%
Time from dosing (Minutes)
*fall ≥ 33% in PI score

Conclusions

- Breakthrough cancer pain is a significant clinical problem, which requires a drug with a fast and reliable onset of action, together with ease of use and high patient acceptability.
- This study presents the first clinical evidence that the nasal delivery of fentanyl using the unique, PecSys-containing, Nasalfent system provides a rapid, consistent and effective treatment for breakthrough pain.
- This is the first study to report clinically meaningful pain relief of breakthrough cancer pain at 5 minutes using a non-injected route of administration.

Nasalfent is an investigational product of Archimedes Development Ltd. For more information please see http://www.archimedespharma.com

References

1. Zeppetella G and Riberio MDC. Pharmacotherapy of cancer-related episodic pain. *Exp Opin Pharmacother* 2003;44(4):493–502.
2. Davis G, Knight AC, Love R *et al*. Nasalfent®, a novel intranasal formulation of fentanyl, is well-tolerated during treatment of breakthrough cancer pain in opioid-tolerant cancer patients. Poster presented at the 10th Congress of the European Association of Palliative Care (EAPC). June 7-9 2007, Budapest, Hungary. Pain 712